The Cardiac Safety of SABER®-Bupivacaine in Patients Undergoing Abdominal Surgery: An Assessment of Holter Monitoring Data From the BESST Trial

Tong J. Gan,[1] Anthony Fossa,[2] Meijian Zhou,[2] David Ellis,[3] Jaymin Shah,[3] Craig Hartrick,[4] Richard Watts[5]

[1]Department of Anesthesiology, Duke University School of Medicine, Durham, North Carolina, USA; [2]iCardiacTechnologies, Rochester, New York, USA; [3]Durect Corp., Cupertino, California, USA; [4]William Beaumont Hospital, Troy, Michigan, USA; [5]Department of Anaesthesia, The Queen Elizabeth Hospital, Woodville, South Australia, Australia

INTRODUCTION

- Bupivacaine is a commonly used local anesthetic for postsurgical pain management and has demonstrated analgesic efficacy in a number of surgical settings[1,2]
 - However, the use of local anesthetics such as bupivacaine is limited by their short duration of action[3]
- To achieve more prolonged postsurgical pain relief, an extended-release formulation of bupivacaine, SABER®-Bupivacaine, has been developed (semiviscous mixture of 12% bupivacaine, which is an amide local anesthetic, sucrose acetate isobutyrate [SAIB], and benzyl alcohol)
- SABER-Bupivacaine is designed to provide analgesia for 72 hours and has been shown to provide pain relief following open hernia repair and shoulder surgery[4,5]
- Numerous studies have been performed to assess the safety profile of bupivacaine
 - Bupivacaine is generally considered safe, but some small trials and animal studies have observed high plasma concentrations of bupivacaine (>2–4 µg/mL) associated with central nervous system and cardiovascular system toxicity[6,7]
- This study evaluated the effect of 5.0 mL (660 mg bupivacaine) SABER-Bupivacaine on Fridericia formula-corrected QT (QTcF) interval

METHODS

Study Design

- The Bupivacaine Effectiveness and Safety SABER Trial (BESST) was an international, multicenter, randomized, double-blind, parallel-group, phase 3 trial that was conducted in patients undergoing elective abdominal surgery
- Eligible patients were enrolled into 1 of 3 cohorts based on the type of surgical procedure and were randomized with a 3:2 allocation ratio to SABER-Bupivacaine or control (bupivacaine hydrochloride [HCl] or SABER-Placebo)
 - Cohort 1, open laparotomy (long incisions; mean, 20 cm): SABER-Bupivacaine 5.0 mL (660 mg) or bupivacaine HCl 30 mL 0.5% solution (150 mg)
 - Cohort 2, laparoscopic cholecystectomy (small incisions; mean, 4 cm): SABER-Bupivacaine 5.0 mL (660 mg) or bupivacaine HCl 30 mL 0.5% solution (150 mg)
 - Cohort 3, laparoscopically assisted colectomy (mean incision, 8–9 cm): SABER-Bupivacaine 5.0 mL (660 mg) or SABER-Placebo 5.0 mL
- SABER-Bupivacaine was instilled directly into the wound, whereas bupivacaine HCl was infiltrated into the peri-incisional tissue with a hypodermic needle
- All patients received medical care given under normal circumstances for the specified elective surgical procedure, including rescue analgesia with opioids

Key Inclusion Criteria

- Men and women at least 18 years old
- Scheduled elective laparotomy, laparoscopic cholecystectomy, or laparoscopically assisted colectomy
- American Society of Anesthesiologists Physical Status I to III
- Body mass index <45 kg/m^2
- Electrocardiogram (ECG) waveform within normal limits or nonspecific ST-T changes; heart rate (HR), 45–100 beats per minute (bpm); PR interval (interval from the beginning of the P wave to the beginning of the QRS complex), ≤220 msec; QRS interval (interval from the beginning of the Q wave to the termination of the S wave), ≤110 msec; and QTcB interval (Bazett-corrected QT interval), <450 msec

Cardiac Exclusion Criteria

- Patients with current or regular use of drugs known to significantly prolong the QTc interval within a period at least 5 times the drug's half-life before day 0
- Patients with atrial fibrillation/flutter or other non-sinus rhythm (including paced rhythm); left bundle branch block; or the following conditions: right bundle branch block in presence of a cardiac disease, clinically significant cardiomyopathy, or myocardial infarction within the past 6 months

Cardiac Assessments and End Points

- Cardiac safety was assessed by digital 12-lead Holter monitoring in all 3 cohorts using the ELA Medical Spiderview Plus device
- During screening, eligible patients underwent a single 24-hour ambulatory Holter recording
- Continuous Holter monitoring was started 1 hour prior to surgery (ie, baseline) and continued until 72 hours after surgery
 - Holter data and blood samples for bupivacaine plasma concentration determination were obtained at baseline and at 0.5, 1, 2, 4, 8, 12, 16, 24, 30, 48, and 72 hours after dosing with the study drug
 - ECGs extracted from the Holter recordings were assessed for changes in HR, interval from the onset of one QRS complex to the onset of the next QRS complex (RR), PR interval, QRS interval, QT interval, QTcB, and QTcF
 - Evaluated using COMPAS software and reviewed by a cardiologist blinded to treatment and sequence
- To detect proarrhythmic events, Holter recordings were analyzed by iCardiac (Rochester, New York) using Global Instrumentation software and transferred to ERT for statistical analysis and reporting
 - Arrhythmia data were analyzed only in cohort 3 (ie, SABER-Bupivacaine vs SABER-Placebo)
- The main ECG end point was SABER-placebo–corrected, baseline-adjusted effects on QTcF (ΔΔQTcF) for SABER-Bupivacaine in cohort 3
- A validated liquid chromatography/tandem mass spectrometry method was used to measure bupivacaine plasma concentrations
- Pharmacokinetic (PK)-pharmacodynamic (PD) modeling was used to evaluate the relationship between bupivacaine plasma concentrations and ΔΔQTcF
- Safety assessments included the monitoring of treatment-emergent adverse events (TEAEs), laboratory findings, wound healing, vital signs, and results of physical examinations

RESULTS

Patients and Disposition

- A total of 305 eligible patients were enrolled into 1 of the 3 cohorts and randomized accordingly
 - Cohort 1: SABER-Bupivacaine (n = 30); bupivacaine HCl (n = 18)
 - Cohort 2: SABER-Bupivacaine (n = 30); bupivacaine HCl (n = 20)
 - Cohort 3: SABER-Bupivacaine (n = 129); SABER-Placebo (n = 78)
- Baseline demographic and clinical characteristics of patients with ECG assessments (n = 300) are presented in **Table 1**
- Baseline (approximately 1 hour prior to surgery/pre-dose) ECG parameters were similar in all cohorts (**Table 1**)

Table 1. Baseline Demographic and Clinical Characteristics of Patients With ECG Assessments Enrolled in BESST (N = 300)

	Cohort 1		Cohort 2		Cohort 3	
	SABER-Bupivacaine	Bupivacaine HCl	SABER-Bupivacaine	Bupivacaine HCl	SABER-Bupivacaine	SABER-Placebo
Demographic data	**n = 29**	**n = 17**	**n = 30**	**n = 20**	**n = 127**	**n = 77**
Age, mean, years	56.4	55.7	44.2	39.5	60.3	58.2
Male, n (%)	17 (58.6)	8 (47.1)	10 (33.3)	6 (30.0)	59 (46.5)	44 (57.1)
BMI, mean, kg/m^2	30.6	27.3	30.8	31.9	29.5	27.5
Baseline ECG parameters	**n = 17**	**n = 12**	**n = 28**	**n = 18**	**n = 103**	**n = 56**
HR, mean (SE), bpm	77.3 (4.2)	82.1 (3.9)	70.8 (2.1)	73.3 (2.2)	71.6 (1.2)	72.3 (1.6)
PR, mean (SE), msec	144.4 (4.6)	151.4 (6.4)	148.2 (4.0)	135.9 (3.9)	151.2 (2.5)[a]	146.5 (2.8)
RR, mean (SE), msec	815.2 (48.3)	749.1 (40.0)	869.5 (26.1)	832.9 (24.8)	863.8 (15.0)	856.8 (22.0)
QRS, mean (SE), msec	106.6 (1.4)	105.6 (1.9)	106.5 (1.1)	105.3 (1.6)	109.4 (1.1)[b]	109.4 (1.1)
QT, mean (SE), msec	393.4 (12.4)	379.3 (10.8)	391.8 (5.7)	384.8 (6.0)	400.0 (3.3)	395.7 (4.4)
QTcF, mean (SE), msec	421.8 (6.4)	418.0 (7.2)	412.0 (3.9)	409.9 (4.3)	421.3 (2.1)	417.9 (2.6)
QTcB, mean (SE), msec	437.9 (4.9)	439.3 (6.8)	422.7 (4.3)	423.3 (4.5)	433.1 (2.2)	430.0 (2.8)

BMI, body mass index; bpm, beats per minute; ECG, electrocardiogram; HR, heart rate; PR, interval from the beginning of the P wave to the beginning of the QRS complex; QRS, interval from the beginning of the Q wave to the termination of the S wave; QT, interval from the beginning of the Q wave to the termination of the T wave; QTcB, QT interval corrected according to Bazett's method; QTcF, QT interval corrected according to Fridericia's method; RR, interval from the onset of one QRS complex to the onset of the next QRS complex; SE, standard error.

[a]Data missing for 2 patients.

[b]Data missing for 1 patient.

Occurrence of TEAEs

- The majority of patients reported ≥1 TEAE
 - The most commonly reported TEAEs were gastrointestinal symptoms (ie, nausea, vomiting, or constipation), which were most likely related to the use of anesthesia, bowel surgery, or opioid use
- Cardiovascular TEAEs were similar between the SABER-Bupivacaine and control groups, with no signs of bupivacaine-related toxicity

Effects on Heart Rate and Electrocardiographic Parameters

- Large procedure-related changes from baseline in HR were seen across all 3 cohorts at 24, 48, and 72 hours post-dose (**Table 2**)
 - In cohort 3, placebo-corrected changes from baseline in HR were not clinically or statistically significant at any time point
- SABER-Bupivacaine did not have any relevant effect on cardiac conduction (the PR and QRS intervals)
 - All groups showed some PR shortening, consistent with the increase in HR
- Likewise, SABER-Bupivacaine did not have a significant effect on cardiac repolarization (**Table 2**)
 - QTcF intervals increased from baseline across all cohorts during the first 4 hours, but shortening was observed at later time points
 - In cohort 3, ΔΔQTcF intervals were <5 msec at all time points, with the exception of 24 hours post-dose when a prolongation of 5.5 msec was observed (**Figure 2**)

Table 2. Changes From Baseline in Heart Rate and Electrocardiographic Parameters for All Cohorts

	Cohort 1		Cohort 2		Cohort 3	
	SABER-Bupivacaine	Bupivacaine HCl	SABER-Bupivacaine	Bupivacaine HCl	SABER-Bupivacaine	SABER-Placebo
ΔHR, mean (90% CI), bpm						
24 hours	17.1 (10.5, 23.7)	3.0 (-5.1, 11.1)	5.6 (3.0, 8.3)	5.1 (1.4, 8.8)	9.0 (6.5, 11.5)	6.7 (4.2, 9.2)
48 hours	15.3 (5.9, 24.7)	14.1 (0.3, 28.0)	7.0 (3.4, 10.6)	3.8 (0.5, 7.0)	14.1 (11.3, 16.9)	11.5 (8.7, 14.3)
72 hours	20.8 (10.2, 31.5)	10.9 (-3.6, 25.3)	7.3 (3.0, 11.6)	4.6 (0.7, 8.5)	11.0 (8.3, 13.6)	9.5 (6.1, 13.0)
ΔPR, mean (90% CI), msec						
24 hours	-14.8 (-21.3, -8.3)	-5.7 (-19.3, 8.0)	-1.0 (-4.8, 2.8)	-1.0 (-6.7, 4.7)	-4.7 (-7.2, -2.2)	-7.8 (-10.8, -4.8)
48 hours	-14.6 (-26.1, -3.1)	-12.9 (-35.9, 10.2)	-7.0 (-11.5, -2.5)	-2.7 (-9.2, 3.7)	-7.3 (-10.1, -4.6)	-10.8 (-14.2, -7.4)
72 hours	-6.7 (-16.6, 3.3)	-15.4 (-31.6, 0.7)	-5.3 (-9.9, -0.8)	-2.8 (-8.6, 2.9)	-10.0 (-12.5, -7.5)	-13.8 (-17.9, -9.7)
ΔQRS, mean (90% CI), msec						
24 hours	-0.1 (-2.4, 2.2)	1.3 (0.0, 2.7)	0.7 (-0.4, 1.7)	-0.5 (-1.8, 0.7)	-0.2 (-1.2, 0.8)	0.0 (-0.8, 0.9)
48 hours	0.3 (-1.9, 2.5)	-1.4 (-3.4, 0.6)	0.2 (-1.0, 1.5)	0.1 (-1.5, 1.8)	-0.6 (-2.0, 0.8)	-0.5 (-1.4, 0.3)
72 hours	0.5 (-2.4, 3.4)	2.0 (-1.6, 5.6)	0.2 (-1.3, 1.7)	-0.6 (-2.3, 1.0)	-0.1 (-1.4, 1.2)	-0.8 (-1.5, 0.0)
ΔQTcF, mean (90% CI), msec						
24 hours	-11.3 (-19.1, -3.5)	-11.2 (-21.1, -1.4)	-0.2 (-5.5, 5.0)	-6.2 (-12.7, 0.2)	-4.2 (-7.0, -1.4)	-9.7 (-13.5, -5.8)
48 hours	-15.4 (-23.1, -7.6)	-24.9 (-34.2, -15.6)	-4.3 (-9.4, 0.9)	-9.1 (-16.0, -2.3)	-14.1 (-16.9, -11.2)	-14.4 (-18.1, -10.6)
72 hours	-13.4 (-22.9, -4.0)	-21.7 (-31.0, -12.4)	-7.5 (-12.6, -2.3)	-9.6 (-16.0, -3.2)	-11.9 (-14.9, -8.9)	-12.4 (-16.5, -8.4)

bpm, beats per minute; CI, confidence interval; HR, heart rate; PR, interval from the beginning of the P wave to the beginning of the QRS complex; QRS, interval from the beginning of the Q wave to the termination of the S wave; QTcF, QT interval corrected according to Fridericia's method.

SABER-Bupivacaine Plasma Concentrations

- Cohorts 1 and 3 had similar concentration profiles, whereas cohort 2 had a shorter T_{max} and a lower C_{max} (**Figure 1**)

Figure 1. SABER-Bupivacaine plasma concentrations.

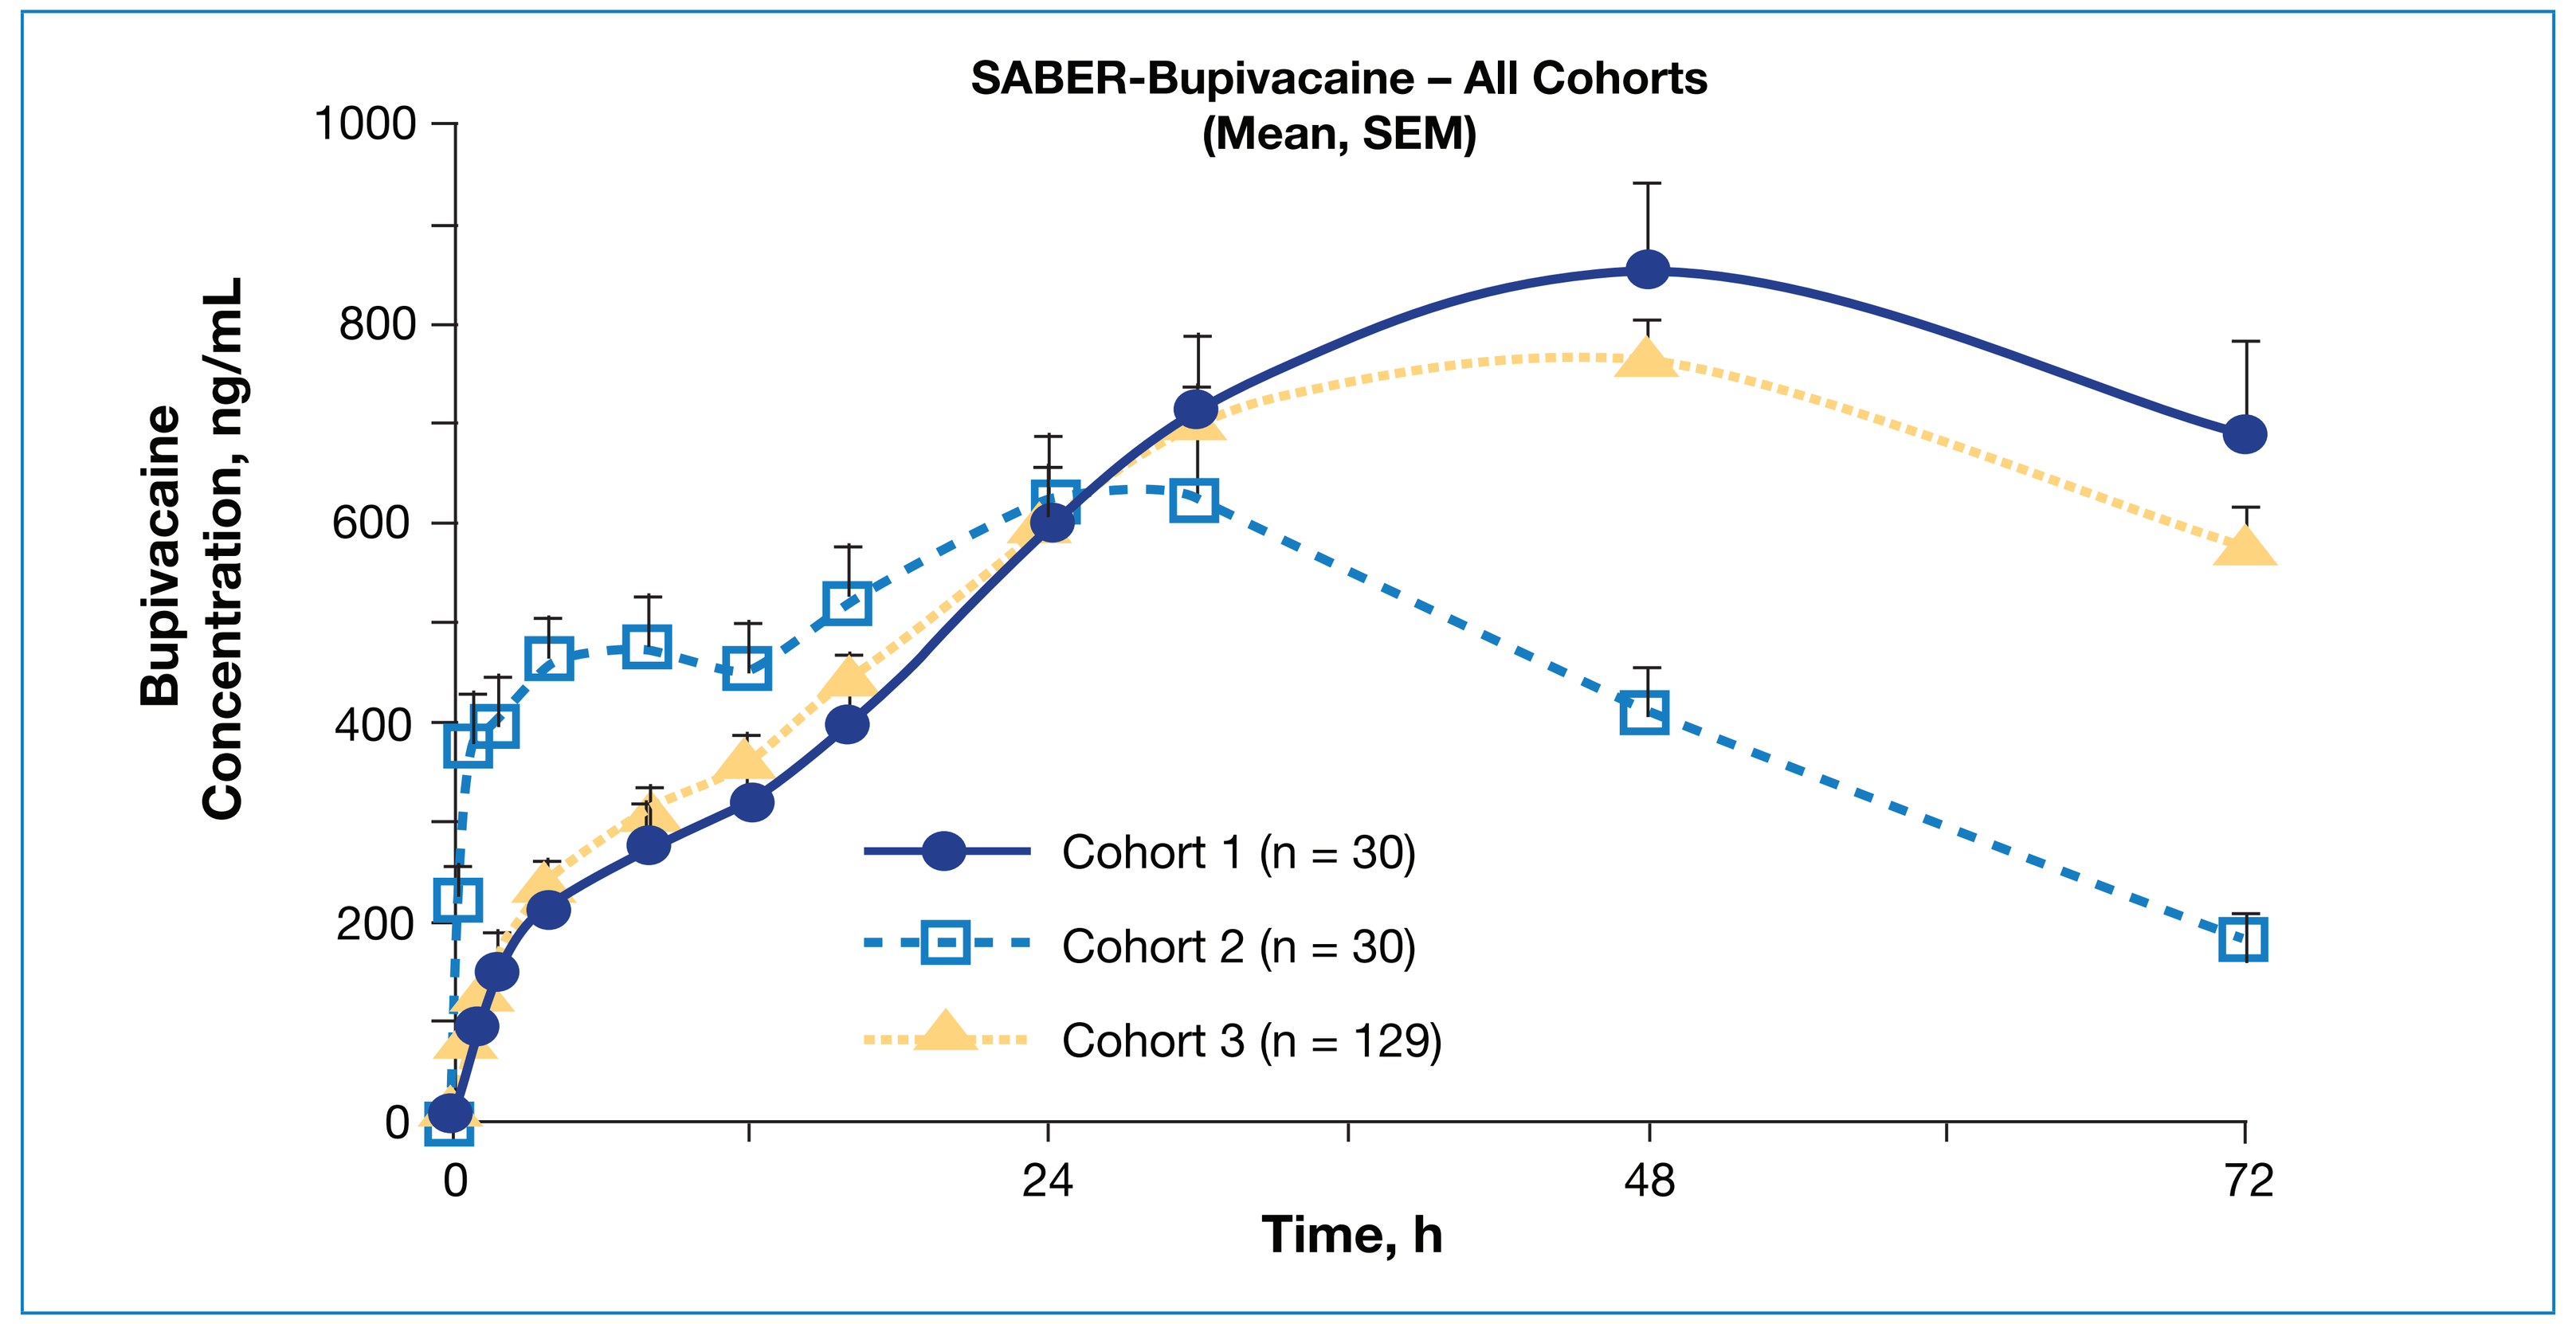
SABER-Bupivacaine – All Cohorts
(Mean, SEM)
Bupivacaine Concentration, ng/mL
1000
800
600
400
200
0
0
24
48
72
Time, h
Cohort 1 (n = 30)
Cohort 2 (n = 30)
Cohort 3 (n = 129)

SEM, standard error of the mean.

Relationship Between QT Prolongation and SABER-Bupivacaine Plasma Concentrations

- No relationship was seen between ΔΔQTcF and observed SABER-Bupivacaine plasma levels (**Figure 2**)
 - Peak plasma levels of SABER-Bupivacaine were seen approximately 48 hours after dosing (cohort 3)
 - In contrast, the largest ΔΔQTcF effect was seen at approximately 24 hours (**Figure 2**)

Figure 2. Relationship between SABER-Bupivacaine plasma concentrations in cohort 3 and placebo-corrected, baseline-adjusted QTcF intervals (ΔΔQTcF).

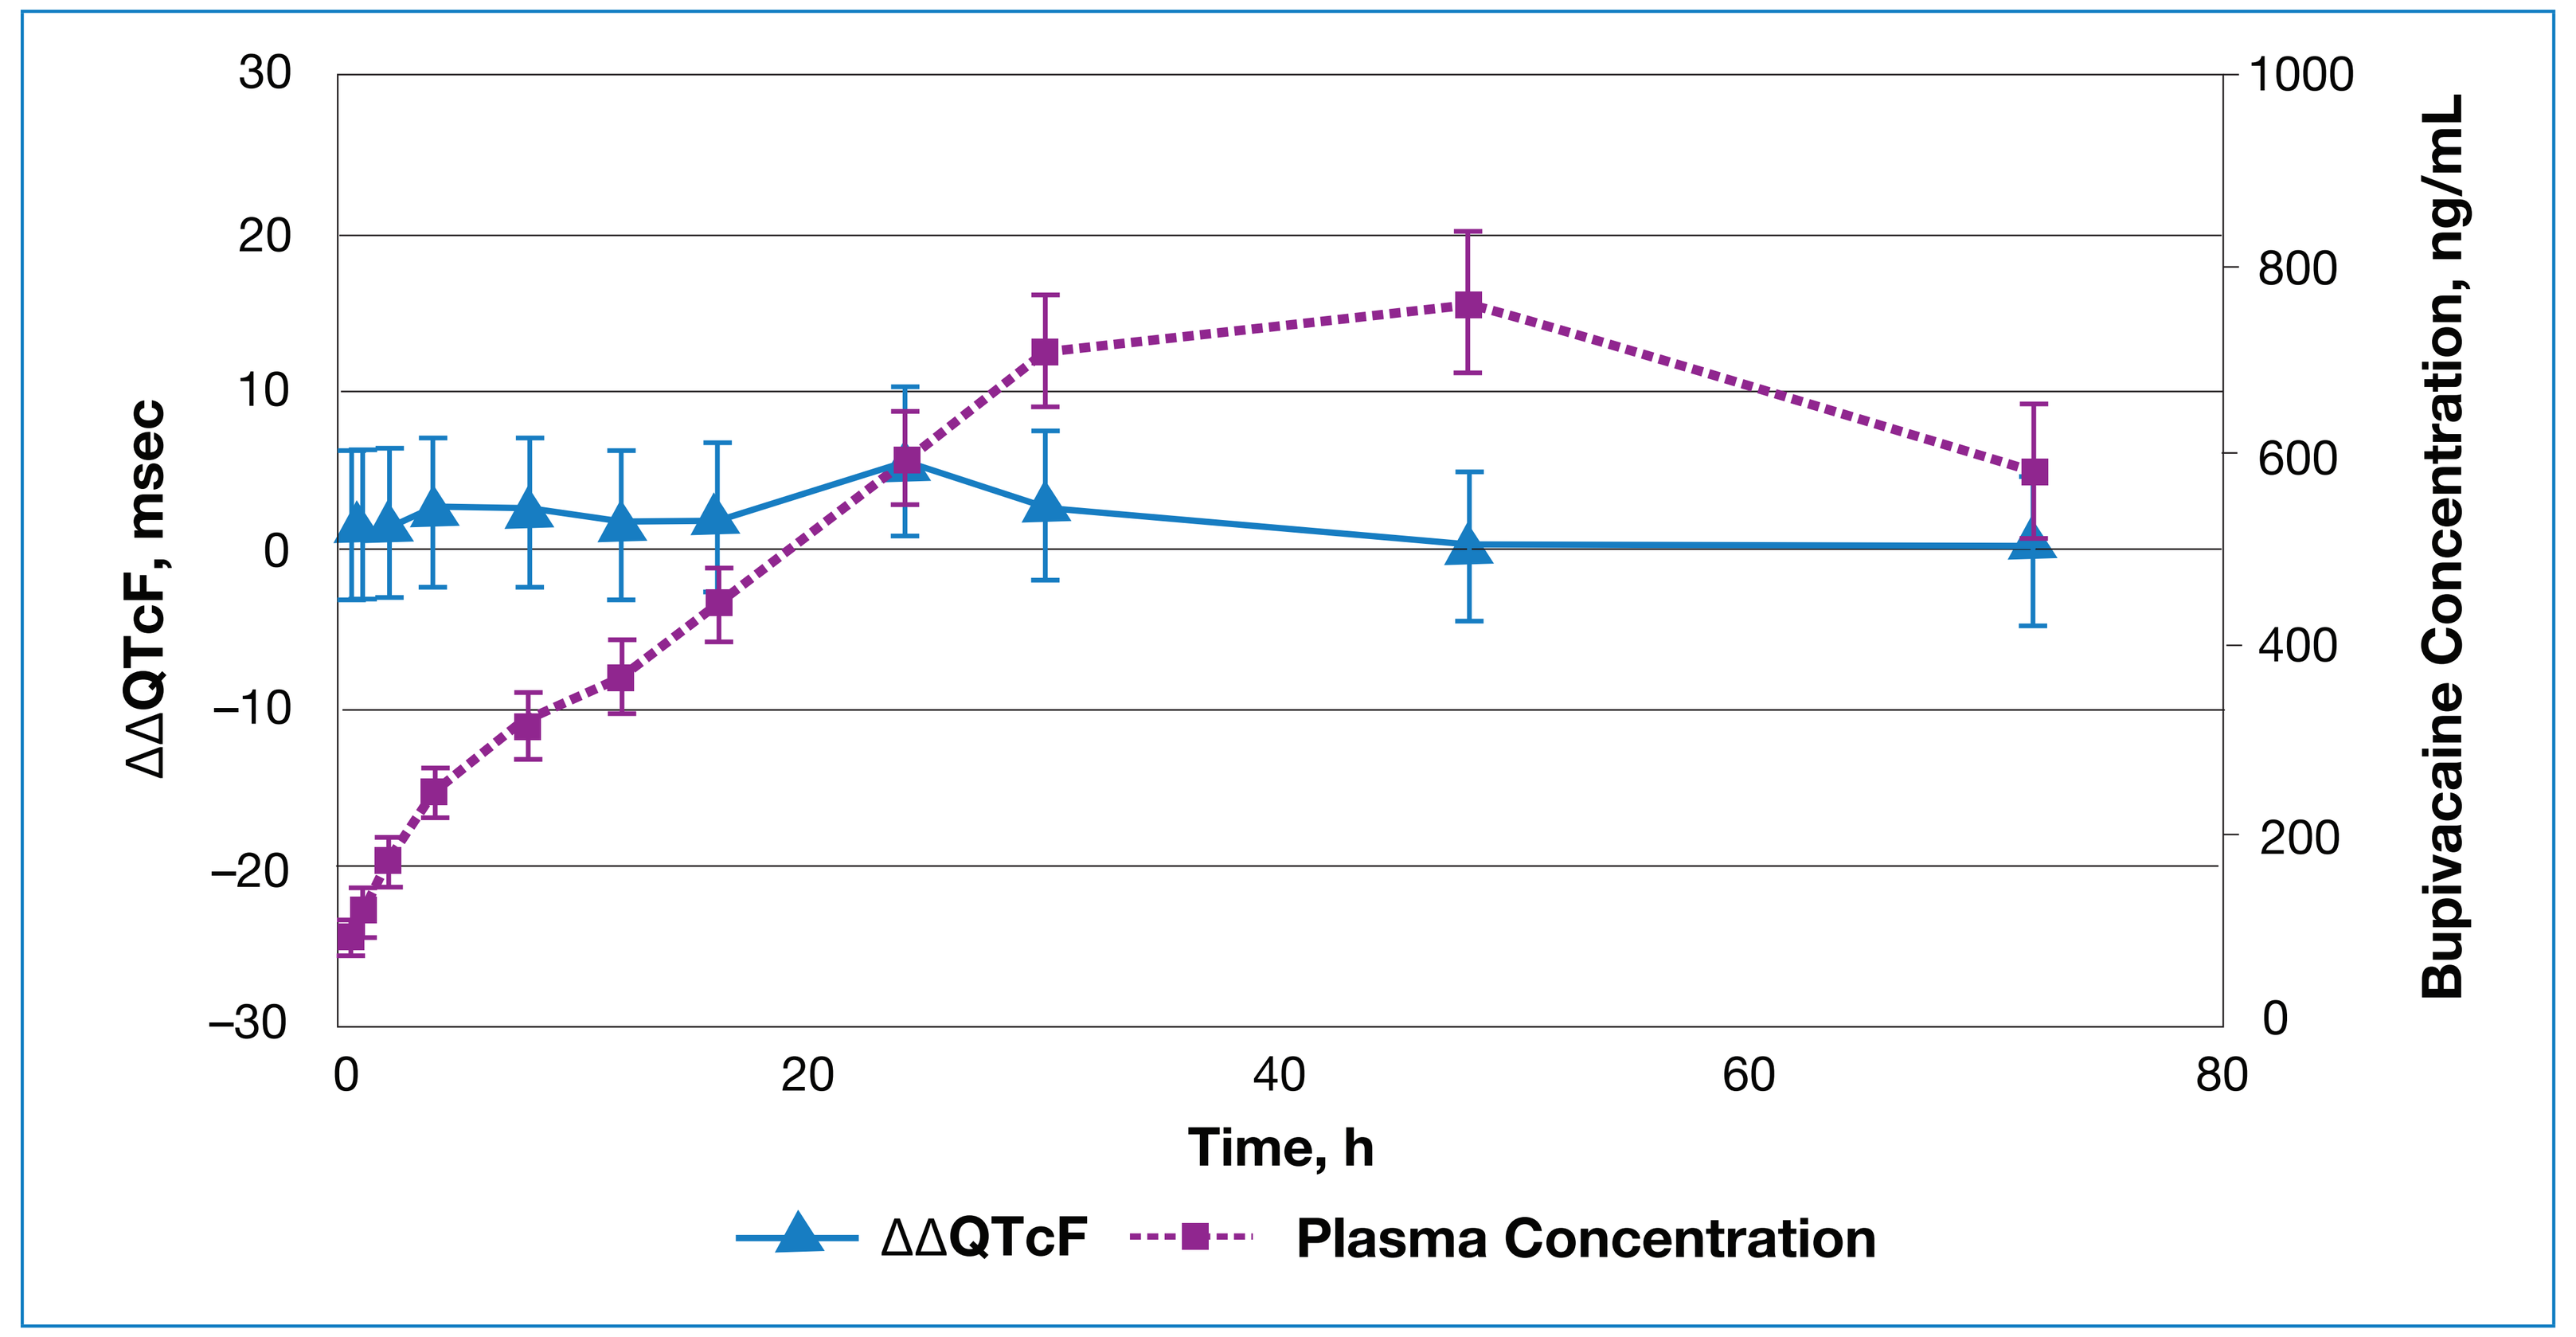
ΔΔQTcF, msec
30
20
10
0
−10
−20
−30
0
20
40
60
80
Time, h
1000
800
600
400
200
0
Bupivacaine Concentration, ng/mL
ΔΔQTcF
Plasma Concentration

Arithmetic means and 90% CI.

- Exposure-response analysis of ΔΔQTcF did not provide evidence of bupivacaine-induced QT prolongation in cohort 3
 - Using the concentration effect model, predicted ΔΔQTcF effect levels at observed peak SABER-Bupivacaine plasma levels were small and below any level of clinical concern (**Figure 3**)
 - The slope of SABER-Bupivacaine was not significantly different from zero

Figure 3. Exposure-response analysis of placebo-corrected, baseline-adjusted QTcF (ΔΔQTcF) in cohort 3.

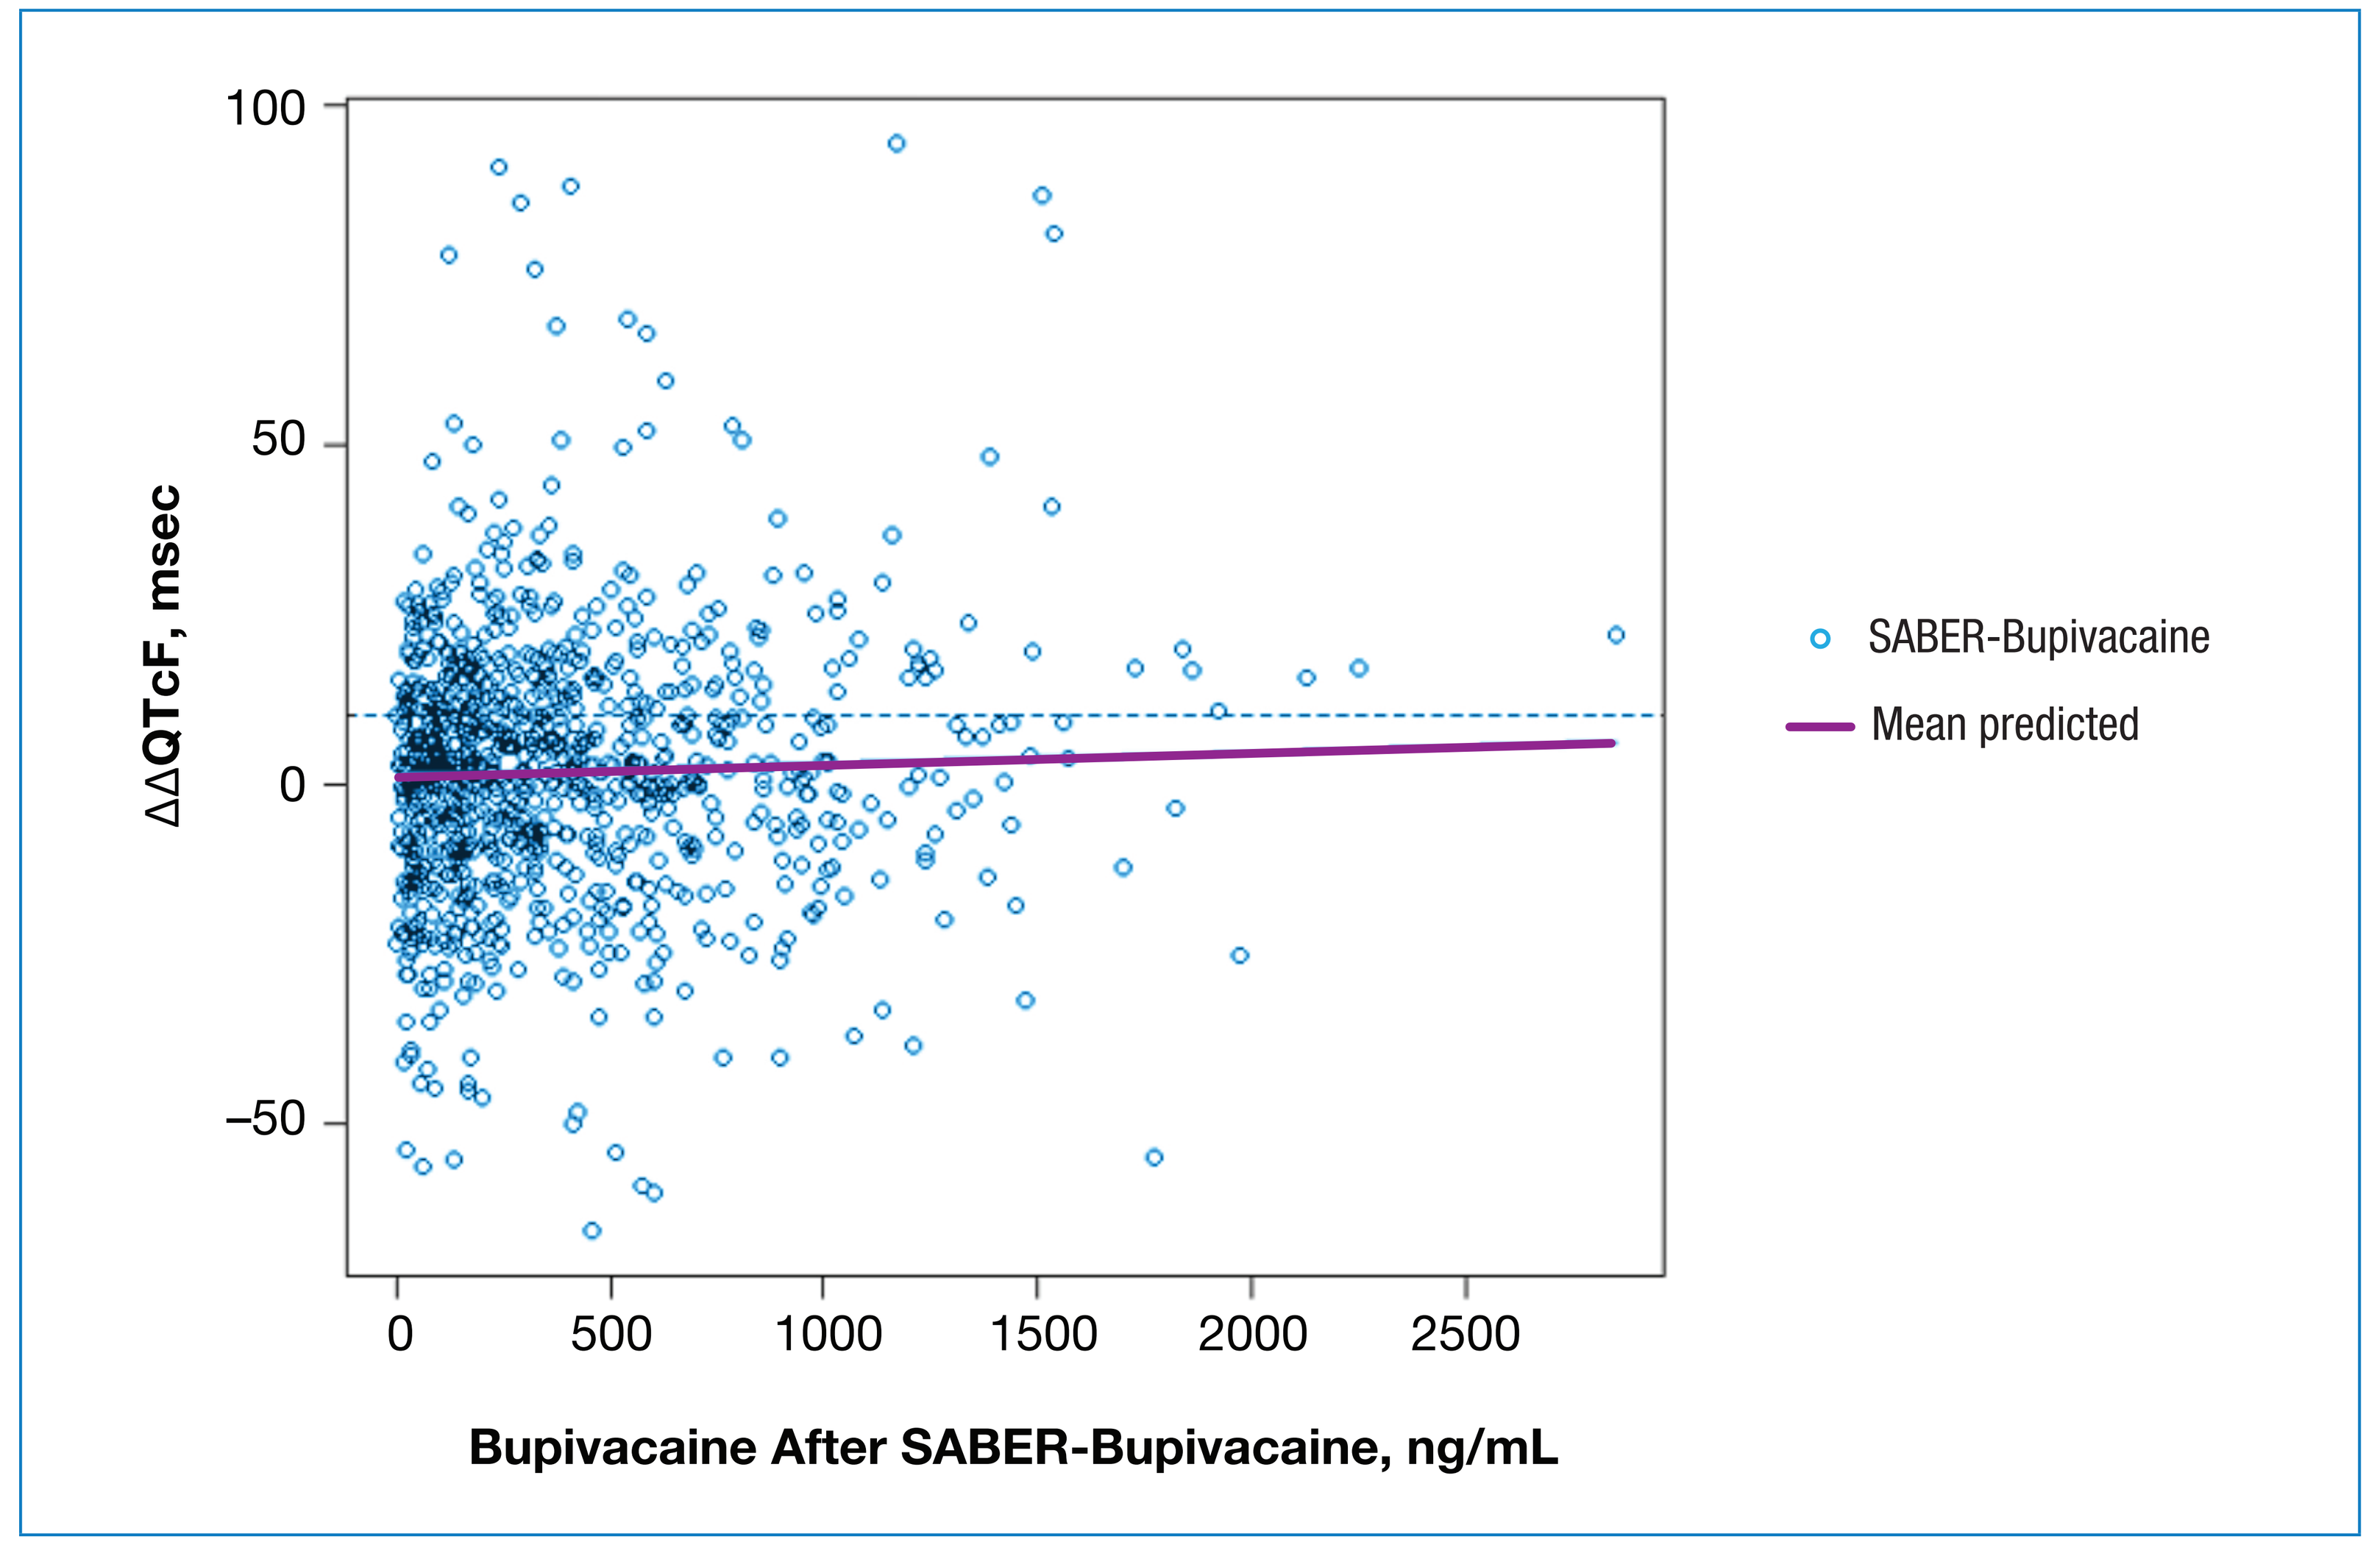
100
50
0
−50
ΔΔQTcF, msec
0
500
1000
1500
2000
2500
Bupivacaine After SABER-Bupivacaine, ng/mL
SABER-Bupivacaine
Mean predicted

Detection of Proarrhythmic Events

- Analyses of the number of supraventricular beats per day and ventricular ectopy frequency showed no clear evidence of clinically significant induction of supraventricular or ventricular arrhythmias
- In addition, the percentages of patients demonstrating any proarrhythmia were not clinically different between SABER-Bupivacaine and SABER-Placebo recipients in cohort 3

CONCLUSIONS

- Holter monitoring showed minimal changes in HR or QT parameters with SABER-Bupivacaine when adjusted for placebo response
- SABER-Bupivacaine enabled extended release of the drug, with maximal plasma concentrations that were below toxic levels
 - This was supported by the PK-PD modeling results of this study, which showed no evidence of a relationship between peak SABER-Bupivacaine plasma levels and QT prolongation
- Analysis of Holter data also showed no evidence of proarrhythmias
- Based on these findings, the administration of SABER-Bupivacaine in patients undergoing abdominal surgery was well tolerated, with no evidence of systemic bupivacaine cardiac toxicity, dose-related QT prolongation, or proarrhythmias
- SABER-Bupivacaine may become an important addition to the treatment options addressing 72-hour pain relief utilizing a local anesthetic, especially in the poorly served 24- to 72-hour postoperative period, while reducing or avoiding opioid use

REFERENCES

1. Moiniche S et al. *Br J Anaesth.* 1998;81:377-83. **2**. American Society of Anesthesiologists Task Force on Acute Pain Management. *Anesthesiology.* 2012;116:248-73. **3.** Collins JB et al. *Can J Plast Surg.* 2013;21:51-3. **4.** Hadj A et al. *ANZ J Surg.* 2012;82:251-7. **5.** Ellis D et al. American Pain Society 32nd Annual Scientific Meeting; May 8, 2013; abstr 423. **6.** Casati A, Putzu M. *Best Pract Res Clin Anaesthesiol.* 2005;19:247-67. **7.** Bergeses SD et al. *Reg Anesth Pain Med.* 2012;37:145-51.

Disclosures

This study was sponsored by DURECT Corporation.

Presented at the International Anesthesia Research Society 2014 Annual Meeting and International Science Symposium; May 17-20, 2014; Montreal, Quebec, Canada

This study was sponsored by the DURECT Corporation.